SYMPTOM RELIEF FROM OAB: WHAT AN "AVERAGE" PATIENT MIGHT EXPECT: DATA FROM A POOLED ANALYSIS OF FESOTERODINE TREATED PATIENTS

151

LABOSSIERE, J. R.[1], FERNET, M.[2], HERSCHORN, S.[1], CARLSSON, M.[3], OELKE, M.[4], WAGG, A.[5]

[1]University of Toronto, ON, Canada [2]Pfizer Canada, Montreal, QC, Canada,
[3] Pfizer Inc. New York, USA, [4] St. Antonius Hospital, Gronau, Germany, [5]University of Alberta, Edmonton AB, Canada

INTRODUCTION & AIMS

Overactive bladder (OAB) affects up to 19% of adults >40 years of age [1]. Severity of symptoms and urgency incontinence drive patients to seek treatment. Data from randomized clinical trials (RCT) support the use of pharmacotherapy but are limited in that the results only describe the average change in index symptoms across the sample of patients entering the study. For newer treatments there is still a dearth of data on what response to treatment an individual patient might expect given their symptoms at presentation, a question often posed to primary care practitioners who are commonly charged with initiating drug therapy. Using pooled trial data, this study sought to characterize patient symptom response following treatment with fesoterodine.

METHODS

This study used pooled data from 6 fixed dose studies, each a parallel, 12 week, double blind RCT to describe the degree of symptom improvement by end of weeks 4, 8 and 12, after exposure to either 4mg, or 8mg of fesoterodine or placebo. Analysis was based on all patients who took at least one dose of assigned study drug and contributed data to at least one baseline and post-baseline efficacy assessment with the baseline value of the outcome variable > 0.

The following measures were used: the proportion of patients achieving a 100% and 50% reduction in

- Urinary urgency episodes/24h **(UUE)**
- Urinary urgency incontinence **(UUI)** episodes /24h (where baseline UUI episodes>0)
- Daytime micturition frequency **(DMF)** /24 h (100% resolution defined as DMF<8/24h)
- Nocturnal Micturition Frequency/24h **(NMF)** (where 100% resolution is defined as N<1)
- **Combined symptoms**, reduction in all of urgency, 24 hr frequency and UUI

RESULTS I

6689 patients were included in all studies. Demographic details and symptom indices at trial entry are shown in Table 1.

Table 1. Baseline characteristics

	Fesoterodine 4mg			Fesoterodine 8mg			Placebo		
	Male	Female	Total	Male	Female	Total	Male	Female	Total
Total sample N	273	1100	1373	576	2687	3263	384	1669	2053
Mean (SD) age	62.1 (12.7)	57.6 (13.2)	58.5 (13.2)	60.0 (14.3)	57.8 (13.0)	58.2 (13.3)	60.8 (13.9)	58.1 (13.1)	58.6 (13.3)
Race (% white)	81.0	85.6	84.7	75.3	82.8	81.5	78.4	83.4	82.5
BMI (m/kg^2) Mean (SD)	28.4 (5.5)	30.0 (7.4)	29.7 (7.1)	28.0 (5.5)	29.5 (6.7)	29.2 (6.5)	28.4 (5.6)	29.5 (7.1)	29.3 (6.8)
DMF/24h mean (SD), N*	9.8 (2.4) 91	10.2 (3.0), 382	10.1 (3.0), 473	9.6 (2.8), 384	9.7 (2.9), 1968	9.7 (2.9), 2352	10.1 (2.9), 264	9.8 (2.9), 1260	9.9 (3.0), 1524
NMF/24h mean (SD), N*	2.5 (2.0), 91	2.0 (1.4), 382	2.1 (1.6), 473	2.4 (1.4), 384	2.1(1.4), 1968	2.1 (1.4), 2352	2.3 (1.4), 264	2.1 (1.4), 1260	2.1 (1.4) 1524
UUE/24h mean (SD), N*	11.7 (4.0), 234	11.2 (4.0), 1029	11.3 (4.0), 1263	10.7 (4.1), 534	10.4 (4.0), 2591	10.4 (4.0), 3125	10.9 (4.4), 334	10.5 (4.1), 1576	10.6 (4.1), 1910
UUI/24h Mean (SD), N*	3.5 (2.4), 234	3.9 (2.7), 1029	3.9 (2.7), 1263	2.9 (2.2), 534	3.3 (2.5), 2591	3.2 (2.5), 3125	3.0 (2.4), 334	3.4 (2.6), 1576	3.3 (2.6), 1910

BM= body Mass Index, DMF=daytime micturition frequency, NMF=nocturnal micturition frequency, UUE= urinary urgency episodes, UUI=urgency incontinence episodes, N* - all patients with UUI>1 at baseline

The proportions of patients experiencing a 50% and 100% reduction in their symptoms at week 4, 8 and 12 in for UUE, UUI, DMF, NMF and all variables according to treatment exposure are shown in Table 2. Figure 1 shows the sample size and proportion of patients experiencing resolution of UUI at each time point. Overall safety of fesoterodine did not differ from that demonstrated in source trials. The most frequent TEAE in the fesoterodine groups were dry mouth and constipation

RESULTS II

		UUE/24h %, (N/sample)	UUI/24h % (N/sample)	DMF/24h % (N/sample)	NMF/24h % (N/sample)	Combined UUE/UUI & DMF/24h % (N/sample)
Week 4						
50% response	**Placebo**	13.3 (257/ 1930)	51.9 (930/ 1792)			
	Fesoterodine 4mg	15.3 (192/ 1257)	**61.7** (712/ 1155)			
	Fesoterodine 8mg	25.0 (762/ 3047)	**69.7** (2043/ 2931)			
100% response	**Placebo**	1.4 (27/ 1930)	23.6 (423/ 1792)	36.6 (574/ 1570)	24.2 (380/ 1570)	0.7% (10/ 1428)
	Fesoterodine 4mg	1.5 (19/ 1257)	**25.7** (297/ 1155)	33.6 (182/ 542)	26.6 (144/ 542)	0.2% (1/ 440)
	Fesoterodine 8mg	2.5 (75/ 3047)	**38.5** (1128/ 3047)	48.9 (1146/ 2343)	27.5 (645/ 2343)	2.1% (47/ 2227)
Week 8						
50% response	**Placebo**	5.2(28/ 539)	45.1 (185/ 410)			
	Fesoterodine 4mg	9.7 (51/ 525)	60.6 (257/ 424)			
	Fesoterodine 8mg	60 (11.3/ 539)	69.0 (303/ 439)			
100% response	**Placebo**	0.6 (3/ 539)	12.2 (50/ 410)	33.2 (179/ 539)	31.4 (169/ 539)	0.2 (1/ 410)
	Fesoterodine 4mg	0.4 (2/ 525)	16.3 (69/ 424)	40.6 (213/ 525)	32.2 (169/ 525)	0.2 (1/ 424)
	Fesoterodine 8mg	0.7 (4/ 539)	23.5 (103/ 439)	47.5 (256/ 539)	37.9 (204/ 539)	0.46 (2/ 439)
Week 12						
50% response	**Placebo**	21.3 (417/ 1955)	63.6 (1156/ 1817)			
	Fesoterodine 4mg	24.8 (319/ 1285)	72.8 (859/ 1179)			
	Fesoterodine 8mg	37.4 (1150/ 3077)	79.8 (2360/ 2959)			
100% response	**Placebo**	3.6 (71/ 1955)	36.1 (655/ 1817)	42.1 (669/ 1589)	29.9 (475/ 1589)	2.9 (42/ 1448)
	Fesoterodine 4mg	3.1 (40/ 1285)	38.9 (459/ 1179)	42.4 (234/ 552)	34.2 (189) 552	0.67 (3/ 446)
	Fesoterodine 8mg	6.4 (198/ 3077)	53.2 (1574/ 2959)	55.0 (1299/ 2361)	32.7 (771/ 2361)	5.22 (117/ 2243)

Table 2. Efficacy variables and responder rates by week and intervention exposure

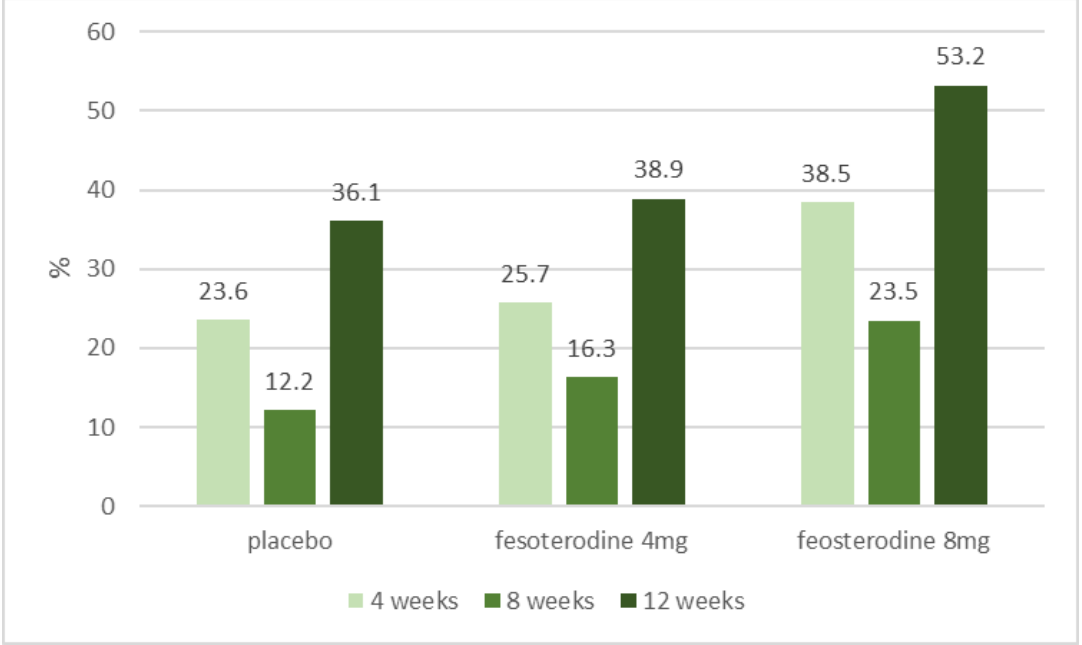
60
50
40
30
20
10
0
%
23.6
12.2
36.1
25.7
16.3
38.9
38.5
23.5
53.2
placebo
fesoterodine 4mg
feosterodine 8mg
4 weeks
8 weeks
12 weeks

	Placebo	Fesoterodine 4mg	Fesoterodine 8mg
Week 4	1792	1155	2931
Week 8	410	424	439
Week 12	1817	1179	2959

Figure 1. Sample size and proportion of patients experiencing resolution of UUI

CONCLUSIONS

- There was an increase in symptom response in patients managed with either 4 or 8 mg of fesoterodine compared to treatment with placebo, reaching 38.9% – 53.2% resolution of UUI at 12 weeks with fesoterodine. At all time points, the decrease in UUE was noticeably smaller than that in UUI and total abolition of UUE was seldom seen.
- The proportion of patients achieving normalization of NMF was not dissimilar to that achieved with placebo.
- The small week 4 sample size may have affected the variability in the estimates of response.
- Total resolution of all symptoms was seldom achieved, regardless of timing or dose of fesoterodine.
- These data, and their time course, provide useful information for patients presenting for treatment in order to convey realistic expectations of symptom relief.

REFERENCES

1. Eur Urol. 2006 Dec;50(6):1306-14

https://congress-download.pfizer.com/ics_2018_philadelphia_ics_2018_453_toviaz_laboissiere_2.html

Statistical support for this study was provided by Pfizer Inc.