Real-World Persistence to Later-Line Therapies Used to Treat Patients With Overactive Bladder

Amy Tung,[1] Jamie Ta,[1] Victor Nguyen,[2] Daniel Mercer[3] Mariana Nelson,[1]

[1]AbbVie, Irvine, CA, USA; [2]Curta Inc, Seattle, WA, USA; [3]Genesis Research, Hoboken, NJ, USA

OBJECTIVE

Evaluate patient characteristics and real-world persistence to later-line treatment among patients with overactive bladder (OAB)

CONCLUSIONS

Among patients with OAB initiating later-line therapy, onabotulinumtoxinA had a higher persistence rate at 14 months post-index date compared with mirabegron and peripheral tibial nerve stimulation (PTNS)

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial.

AbbVie funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA.

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: Amy Tung, Jamie Ta, and Mariana Nelson are employees of AbbVie and may hold AbbVie stock. Victor Nguyen is an employee of Curta Inc, and Danial Mercer is an employee of Genesis Research, which both provide consulting services to AbbVie.

This data was originally presented at the 2022 Academy of Managed Care Pharmacy Annual Meeting (AMCP 2022).

For additional information or to obtain a PDF of this poster

Scan QR code or use the following link to download an electronic version of this presentation and other ICS 2022 scientific presentations: https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=446

QR Code expiration: 10 August 2023

To submit a medical question, please visit www.abbviemedinfo.com

References

1. Haylen BT, et al. *Int Urogynecol J.* 2010;21(1):5-26.
2. Gormley EA, et al. Diagnosis and Treatment of Overactive Bladder (Non-Neurogenic) in Adults: AUA/SUFU Guideline. 2019. Accessed February 10, 2022. https://www.auanet.org/guidelines/non-oncology-guidelines/incontinence.
3. Sussman D, et al. *Int J Clin Pract.* 2017;71(3-4):e12824.
4. Nitti VW, et al. *J Urology.* 2016;196:791-800.
5. Eldred-Evans D, et al. *Ther Adv Urol.* 2017;9(1):3-10.
6. Hamid R, et al. *Int Urogynecol J.* 2021;32:65-74.
7. McCammon K, et al. *Female Pelvic Med Reconstr Surg.* 2021;27:450-456.

INTRODUCTION

- OAB is a chronic condition characterized by symptoms of urinary urgency, often accompanied by frequency and nocturia, with or without urinary incontinence[1]
- Treatments for OAB include first-line behavioral therapies, second-line pharmacologic management with oral therapies, and third-line therapies including intradetrusor injection with onabotulinumtoxinA, peripheral tibial nerve stimulation (PTNS), or sacral nerve stimulation[2]
- Anticholinergics have previously shown high rates of treatment discontinuation over 12 months (84%),[3] highlighting the need for other treatment options
- Currently there is little available evidence regarding rates of real-world persistence with later-line therapies such as onabotulinumtoxinA, beta-3 agonists (mirabegron), and PTNS

METHODS

Study Design

IBM MarketScan Commercial and Medicare Supplemental Database Retrospective Cohort Analysis Study Period

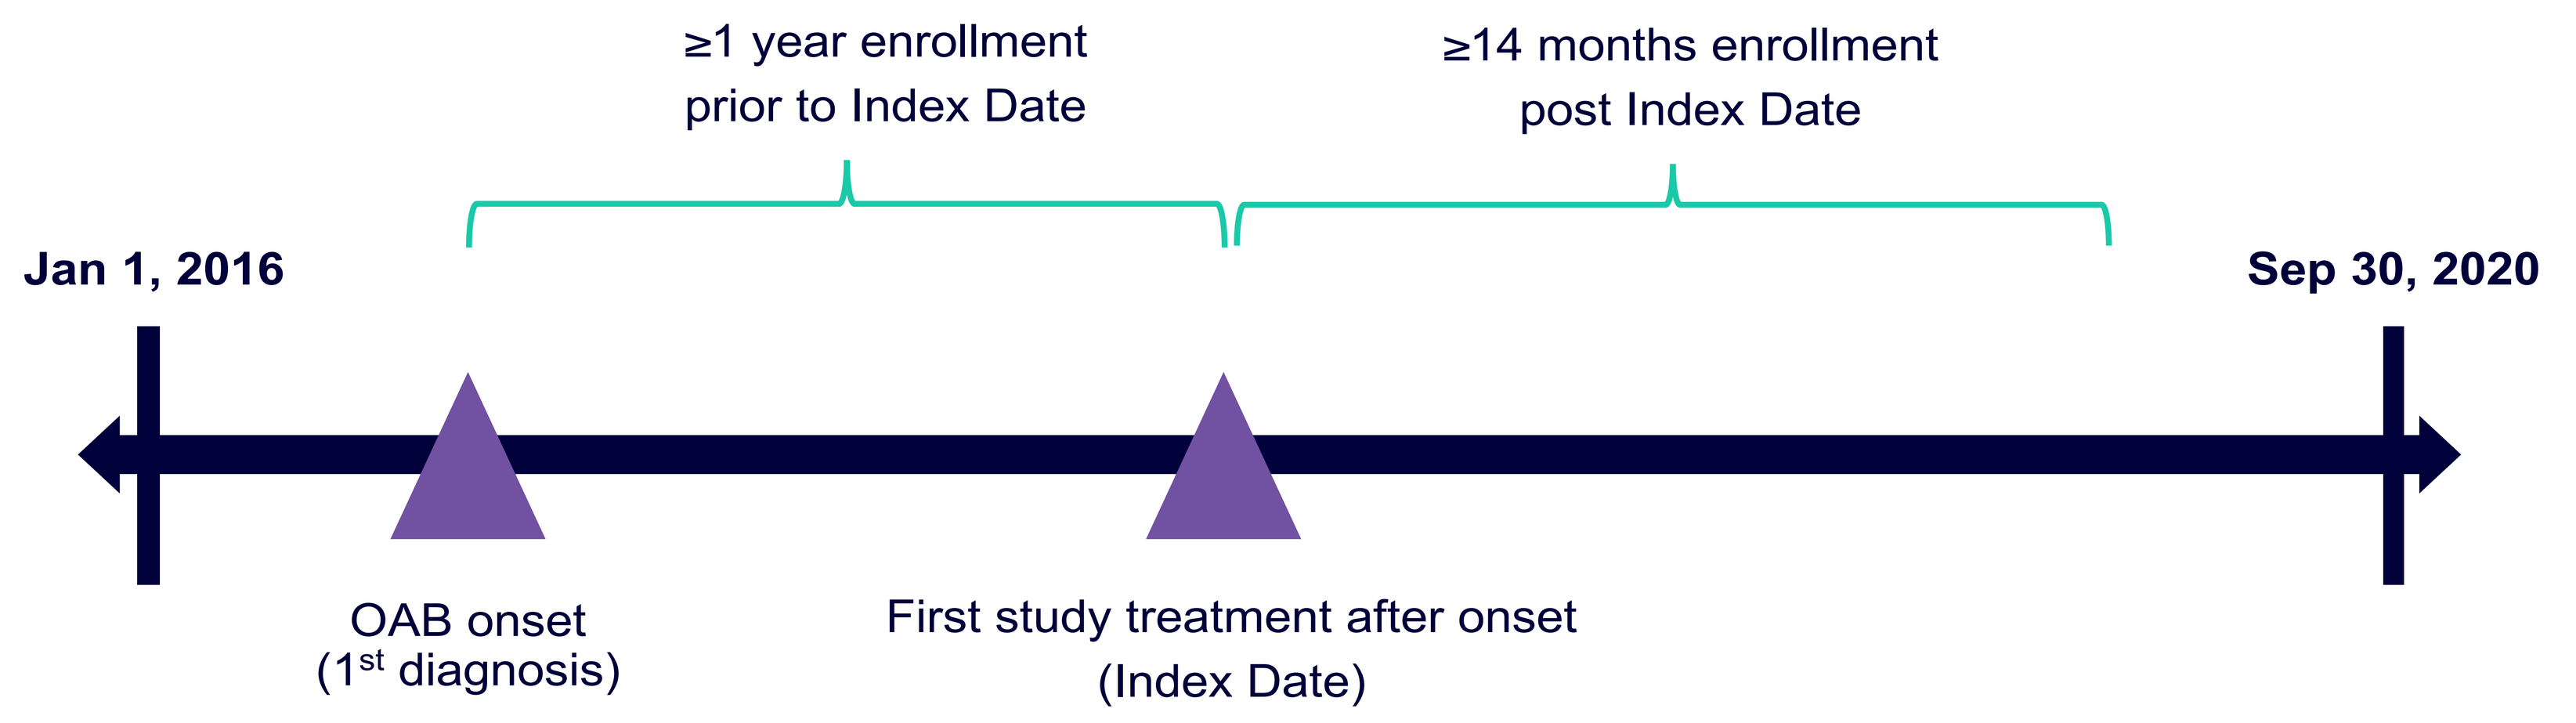
≥1 year enrollment prior to Index Date
≥14 months enrollment post Index Date
Jan 1, 2016
Sep 30, 2020
OAB onset (1st diagnosis)
First study treatment after onset (Index Date)

- A 14-month post-index window was selected for persistence follow-up, allowing for 2 or more onabotulinumtoxinA injections in the 14 months, considering that in clinical practice, injections for OAB are administered on average every 7 months (with a high degree of variability)[4-7]
- Cohort Attrition Index can be found in the supplemental material (QR code)

Inclusion Criteria

- Diagnosis of OAB on at least 1 inpatient or 2 outpatient medical claims ≥60 days apart between January 1, 2016 and September 30, 2019
- ≥1 claims for onabotulinumtoxinA, mirabegron, and PTNS with OAB diagnosis on or after OAB onset (earliest claim = index date)
- Adults ≥18 years at index date
- Continuous enrollment in medical and pharmacy benefits in the 1 year prior to index date and at least 14 months after index date with a gap no greater than 30 days

Exclusion Criteria

- Evidence of onabotulinumtoxinA, mirabegron, or PTNS use any time prior to index during study period
- Use of toxins other than onabotulinumtoxinA during the study period
- Patients with at least 1 inpatient OR at least 2 outpatient non-diagnostic medical claims (≥30 days apart) with a diagnosis of neurogenic bladder during the study period

Outcomes & Definitions

- For each patient cohort, outcomes included:
 - 12-month pre-index demographics and clinical characteristics
 - 14-month post-index rate of persistence to onabotulinumtoxinA, mirabegron, and PTNS
- A sensitivity analysis was performed with alternate persistence definitions for mirabegron and PTNS that were more similar to how persistence was defined for onabotulinumtoxinA

Therapy	Definitions of Persistence With Treatment (Primary Analysis)	Definitions of Persistence for Sensitivity Analysis
OnabotulinumtoxinA	≥2 injections during follow-up period	---
Mirabegron	Continuous days' supply coverage with <30-day gap during follow-up period	≥7 prescription fills during follow-up period
PTNS	≥12 treatments in first 12 weeks followed by ≥1 treatment per month, with <30-day gap during remaining follow-up period	≥12 treatments in first 12 weeks, followed by ≥6 treatments during remaining follow-up period

PTNS, percutaneous tibial nerve stimulation.

RESULTS

Characteristic	OnabotulinumtoxinA N=698	Mirabegron N=9249	PTNS N=314
Age, mean (SD)	61.1 (13.77)	62.7 (15.23)	77.8 (7.31)
Sex, %			
Female	87	72	71
Male	13	29	29
Region, %			
North Central	25.1	15.1	21.0
Northeast	15.8	27.1	26.4
South	34.5	32.3	35.4
West	10.6	6.9	16.9
Unknown	14.2	18.6	0.3
Plan type, %			
Commercial	68	61	3
Medicare	32	39	97
Baseline CCI, mean (SD)	1.5 (1.91)	1.6 (1.95)	2.1 (2.05)
CCI score, %			
0	42	40	28
1	23	21	21
2	14	14	19
3	22	24	33

CCI, Charlson Comorbidity Index; PTNS, percutaneous tibial nerve stimulation; SD, standard deviation.

- Mean baseline age and Charlson Comorbidity Index (CCI) scores were higher in the PTNS group; the majority in this group were covered by Medicare rather than commercial insurance
- Persistence rate at 14 months post index date was highest in the onabotulinumtoxinA group (P<0.0001) in both the primary analysis and in a sensitivity analysis using more permissive definitions of persistence for mirabegron and PTNS groups

14-Month Persistence Rates by Treatment Cohort

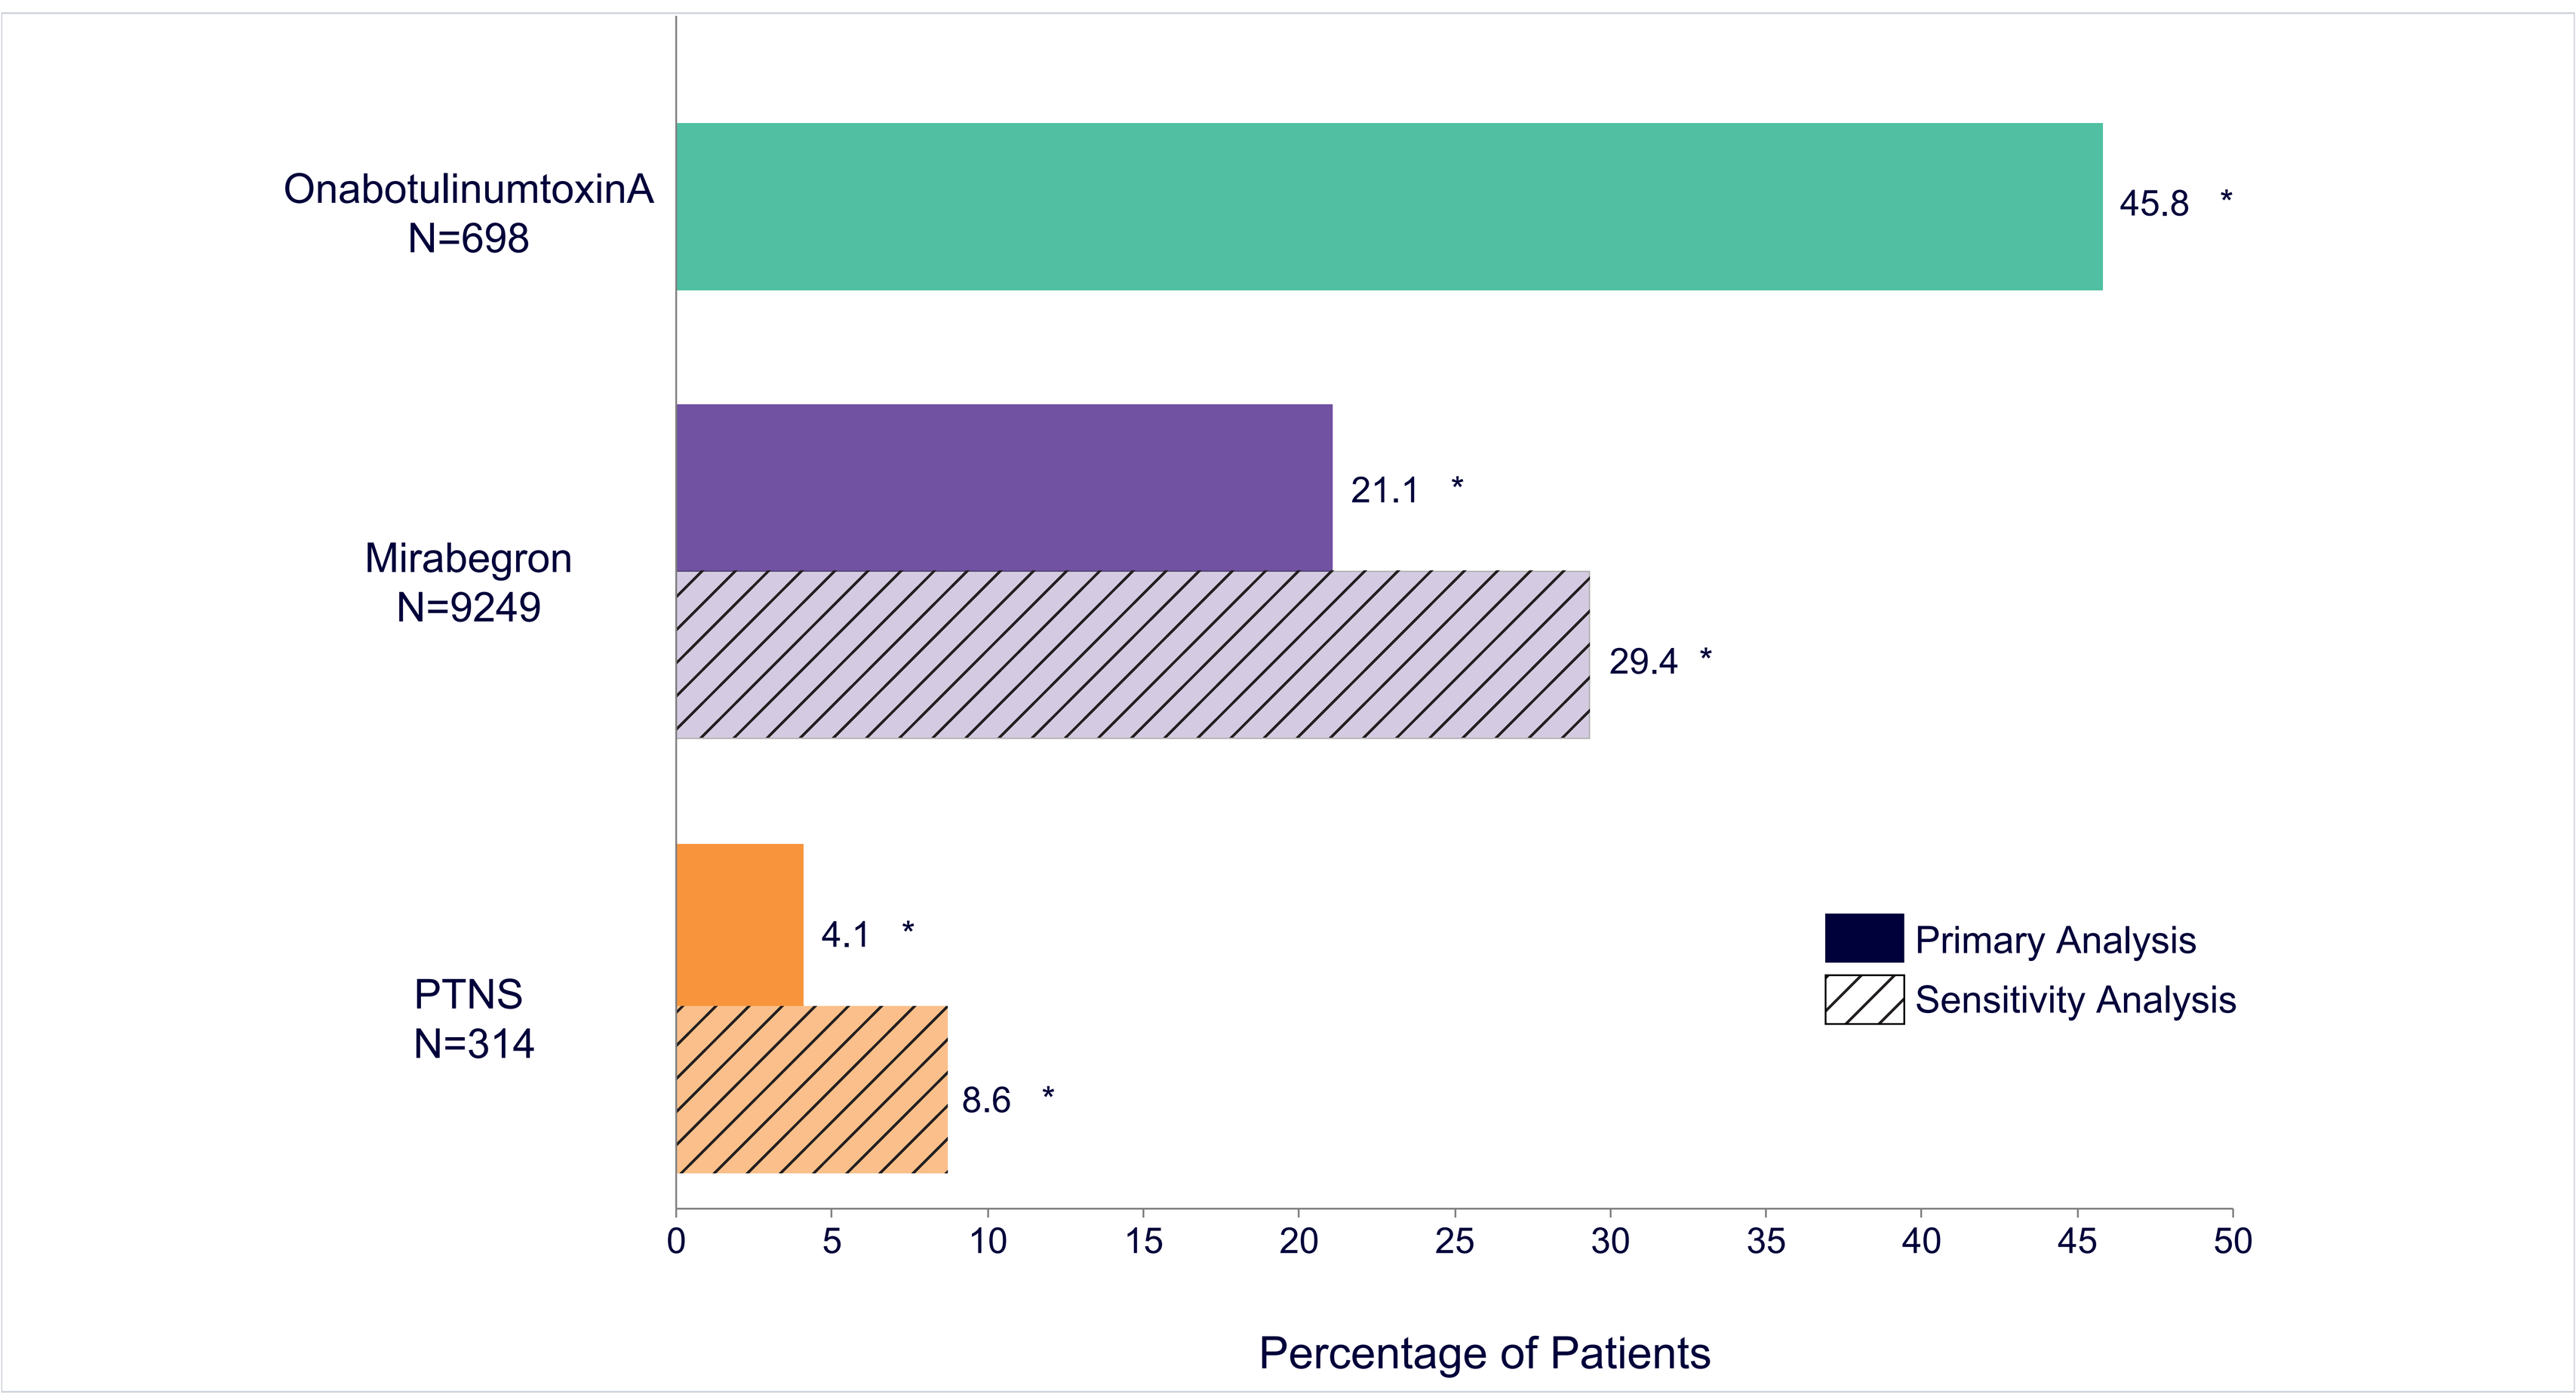
OnabotulinumtoxinA N=698
45.8 *
Mirabegron N=9249
21.1 *
29.4 *
PTNS N=314
4.1 *
8.6 *
Primary Analysis
Sensitivity Analysis
0 5 10 15 20 25 30 35 40 45 50
Percentage of Patients

P<0.0001 for between-group differences.
PTNS, percutaneous tibial nerve stimulation.

Presented at the International Continence Society (ICS) 2022 Scientific Meeting, September 7-10, 2022, Vienna, Austria