SHORT-TERM RESULTS OF CURCUMIN, QUERCETIN, HYALURONIC ACID AND CHONDROITIN SULFATE (IALURIL SOFT GEL®) IN THE MANAGEMENT OF CHRONIC PROSTATITIS/PRIMARY PROSTATE PAIN SYNDROME: A SINGLE-CENTER PROSPECTIVE STUDY.

Iacovelli V, Carilli M, Bertolo R, Vittori M, Petta F, Signoretti M, Maiorino F, Filippi B, Bove P.

Urology Unit, San Carlo di Nancy General Hospital – GVM Care and Research – Rome, Italy

Introduction

Management of Chronic Prostatitis/Primary Prostate Pain Syndrome (CP/PPPS) is challenging and characterized by conflicting results.
The philosophy for the management of chronic pelvic pain is based on a bio-psychosocial model. This is a holistic approach with patients' active involvement.

Figure 1. Phenotyping of pelvic pain - UPOINT classification

Phenotyping	Assessment
Urology	Urinary flow, micturition diary, cystoscopy, ultrasound, uroflowmetry.
Psychology	Anxiety about pain, depression and loss of function, history of negative sexual experiences.
Organ specific	Ask for gynaecological, gastro-intestinal, ano-rectal, sexological complaints. Gynaecological examination, rectal examination.
Infection	Semen culture and urine culture, vaginal swab, stool culture.
Neurological	Ask for neurological complaints (sensory loss, dysaesthesia). Neurological testing during physical examination: sensory problems, sacral reflexes and muscular function.
Tender muscle	Palpation of the pelvic floor muscles, the abdominal muscles and the gluteal muscles.
Sexological	Erectile function, ejaculatory function, post-orgasmic pain.

Aim of the study

This study aimed to evaluate the efficacy of a food supplement based on Curcumin, Quercetin, Hyaluronic Acid and Chondroitin Sulfate (Ialuril Soft Gel®) in CP/PPPS treatment.

Study Design, Methods and Materials

This is a **single cohort pilot study**.

Data of consecutive male patients who referred to our Institution for CP/PPPS were prospectively collected between Oct-2022 and Jan-2023.

Exclusion criteria:
- maximum flow rate < 15 ml/s,
- post-void residual > 150 ml
- previous prostate surgery were excluded.

Variables including age, body mass index (BMI), prostate volume (PVol) and serum prostate specific antigen (PSA) were collected. Patients were asked to fulfil standardized **questionnaires** to assess severity of pain, lower urinary tract symptoms (LUTS) and erectile function, including
- Symptom Severity Index (SSI),
- Symptom Frequency Questionnaire (SFQ),
- National Institutes of Health Chronic Prostatitis Symptom Index (NHI/CPSI) (pain and LUTS domains),
- International Prostate Symptom Score (IPSS) and Quality of Life score (IPSS-QoL), and
- International Index of Erectile Function (IIEF-5).

Changes in standardized questionnaires were evaluated at baseline, 30- and 90-days after enrolment.

Patients were administered with **Ialuril Soft Gel**® (curcumin 200 mg, quercitin 200 mg, hyaluronic acid 100 mg, chondroitin sulfate 200 mg) 2 gel caps **once a day** for 60 days.

Table 1. Ialuril Soft Gel composition.

	2 gel caps dose (mg)
Curcumin	200
Quercitin	200
Hyaluronic Acid	100
Chondroitin sulfate	200

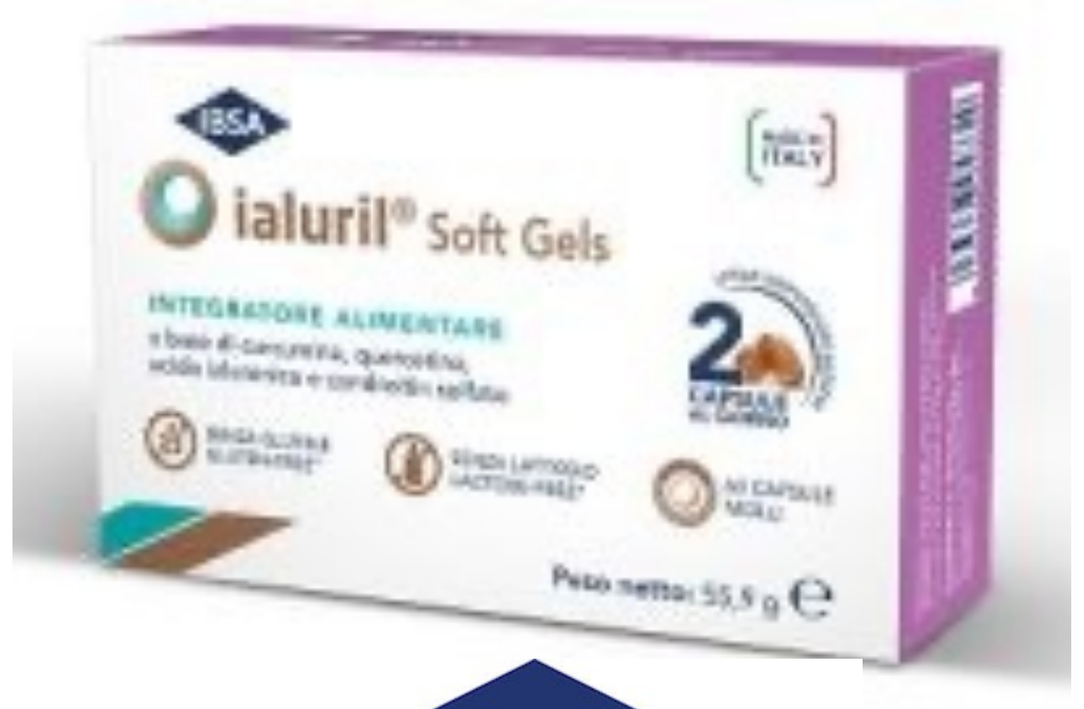
IBSA
ialuril® Soft Gels
INTEGRATORE ALIMENTARE

Results

Twenty patients were analyzed. Baseline features were age [median 50 years (IQR 46-51)], BMI [24.2 (22.8-28.3)], PVol [42 ml (30-52)], PSA [1.0 ng/ml (0.8-1.8)], SSI [54 (27-55)], SFQ [20 (13-26)], NHI/CPSI [pain domain 10 (8-13); LUTS domain 7 (4-8)], IPSS [13 (10-18)], IPSS-QoL [3 (2-3)] and IIEF-5 [17 (14-22)].

SSI and SFQ scores showed statistically significant differences both at 30- [SSI 45 (22-49), p < 0.001; SFQ 16 (11-20), p = 0.035] and 90-days [SSI 33 (17-42), p < 0.001; SFQ 13 (9-16), p < 0.001].

IPSS and NHI/CPSI pain and LUTS domains showed statistically significant differences at 30-days [IPSS 12 (10-13), p = 0.002; NHI/CPSI pain domain 6 (5-9), p < 0.001; NHI/CPSI LUTS domain 7 (2-9), p = 0.033, respectively], but not at 90-days (all p > 0.05).

IPSS-QoL and IIEF-5 scores did not report any statistically significant difference during follow-up (p=0.12 and p=0.23, respectively).

No adverse events were recorded.

Table 2. Validated questionnaires results at baseline, 30 days of treatment, 90 days after starting treatment (= 30 days after end of treatment).

	Baseline	30 days of treatment		90 days after starting treatment	
SSI	54 (27-55)	45 (22-49)	**p < 0.001**	33 (17-42)	**p < 0.001**
SFQ	20 (13-26)	16 (11-20)	**p = 0.035**	13 (9-16)	**p < 0.001**
NHI/CPSI	pain domain 10 (8-13) LUTS domain 7 (4-8)	pain domain 6 (5-9) LUTS domain 7 (2-9)	**p < 0.001** **p = 0.033**	pain domain 7 (5-11) LUTS domain 8 (3-9)	p = 0.2 p = 0.33
IPSS	13 (10-18)	12 (10-13)	**p = 0.002**	11 (10-13)	p=0.6
IPSS-QoL	3 (2-3)	3.2 (2-4)	p=0.12	3.3 (2-4)	p=0.15
IIEF-5	17 (14-22)	18 (13-21)	p=0.23	18 (14-22)	p=0.3

Interpretation of results

PPPS is a complex condition which needs of a multimodal and phenotypically directed treatment options.

The food supplementation based on Curcumin, Quercetin, Hyaluronic Acid and Chondroitin Sulfate may positively impact pain, QoL and urinary symptoms in men.

Conclusions

Ialuril Soft Gel® seems to be effective in reducing the severity of pain and urinary symptoms in CP/PPPS patients at short-term follow-up.
Further powered studies are mandatory to better comprehend the role of Ialuril Soft Gel ® in the management of CP/PPPS.

References

1. EAU Guidelines on Chronic Pelvic Pain. Edn. presented at the EAU Annual Congress Milan 2023. ISBN 978-94-92671-19-6.
2. Nickel JC, Johnston B, Downey J, et al. Pentosan polysulfate therapy for chronic nonbacterial prostatitis (chronic pelvic pain syndrome category IIIA): a prospective multicenter clinical trial. Urology. 2000 Sep 1;56(3):413-7.

Disclosure

Funding This study has been supported by IBSA - Caring Innovation
Clinical Trial, No Subjects, Human Ethics Committee: Comitato Etico Lazio1
Helsinki Yes Informed Consent Yes

ICS 2023 TORONTO